Discovery of a potent KRAS macromolecule degrader specifically targeting tumours with mutant KRAS

Nicolas Bery[a,d], Ami Miller[a,e], Sandrine Leggb, Judit Debreczeni[c], Jason Breed[c], Kevin Embrey[c], Christopher Stubbs[c], Paulina Kolasinska-Zwierz[b], Nathalie Barrett[b], Rose Marwood[b], Jo Watson[b], Jon Tart[c], Ross Overman[c], Christopher Phillips[c], Ralph Minter[b], Terry Rabbitts[a,e]

a Weatherall Institute of Molecular Medicine, MRC Molecular Haematology Unit, University of Oxford, John Radcliffe Hospital, Oxford, OX3 9DS, UK
b Antibody Discovery and Protein Engineering, R&D BioPharmaceuticals, Milstein Building Granta Park, Cambridge, CB21 6GH, UK
c Discovery Sciences, R&D BioPharmaceuticals, AstraZeneca, Darwin Building, Cambridge Science Park, Milton Road, Cambridge, CB4 0WG, UK
d Centre de Recherche en Cancérologie de Toulouse, UMR 1037, INSERM - Université Toulouse III – ERL5294 CNRS, France
e Institute of Cancer Research, Division of Cancer Therapeutics, 15 Cotswold Road, Sutton, London, SM2 5NG, UK

Tumour-associated KRAS mutations are the most prevalent in the three RAS-family isoforms and involve many different amino-acids. Therefore, molecules able to interfere with mutant KRAS protein are potentially important for wide-ranging tumour therapy. We describe here (1) the selection and characterisation of a KRAS-specific antibody mimetic (Designed Ankyrin Repeat Protein, DARPin K19) and (2) the engineering of this DARPin into RAS degraders based on protein macromolecules fused to specific E3 ligases. The KRAS-specific DARPin K19 fused to the VHL E3 ligase is compared to a pan-RAS intracellular single domain antibody (iDAb RAS) fused to the UBOX domain of the CHIP E3 ligase. We demonstrate that while the KRAS-specific DARPin degrader induces specific proteolysis of both mutant and wild type KRAS, it only inhibits proliferation of cancer cells expressing mutant KRAS *in vitro* and *in vivo*. Pan-RAS protein degradation, however, impairs proliferation irrespective of the RAS mutation. These data show that specific KRAS degradation is an important therapeutic strategy to affect tumours expressing any of the range of KRAS mutations.

- 100% conserved between K/N/HRAS isoforms
- GDP/GTP selectivity
- Inhibitors binding +++

- 90% conserved
- Isoform specificity?

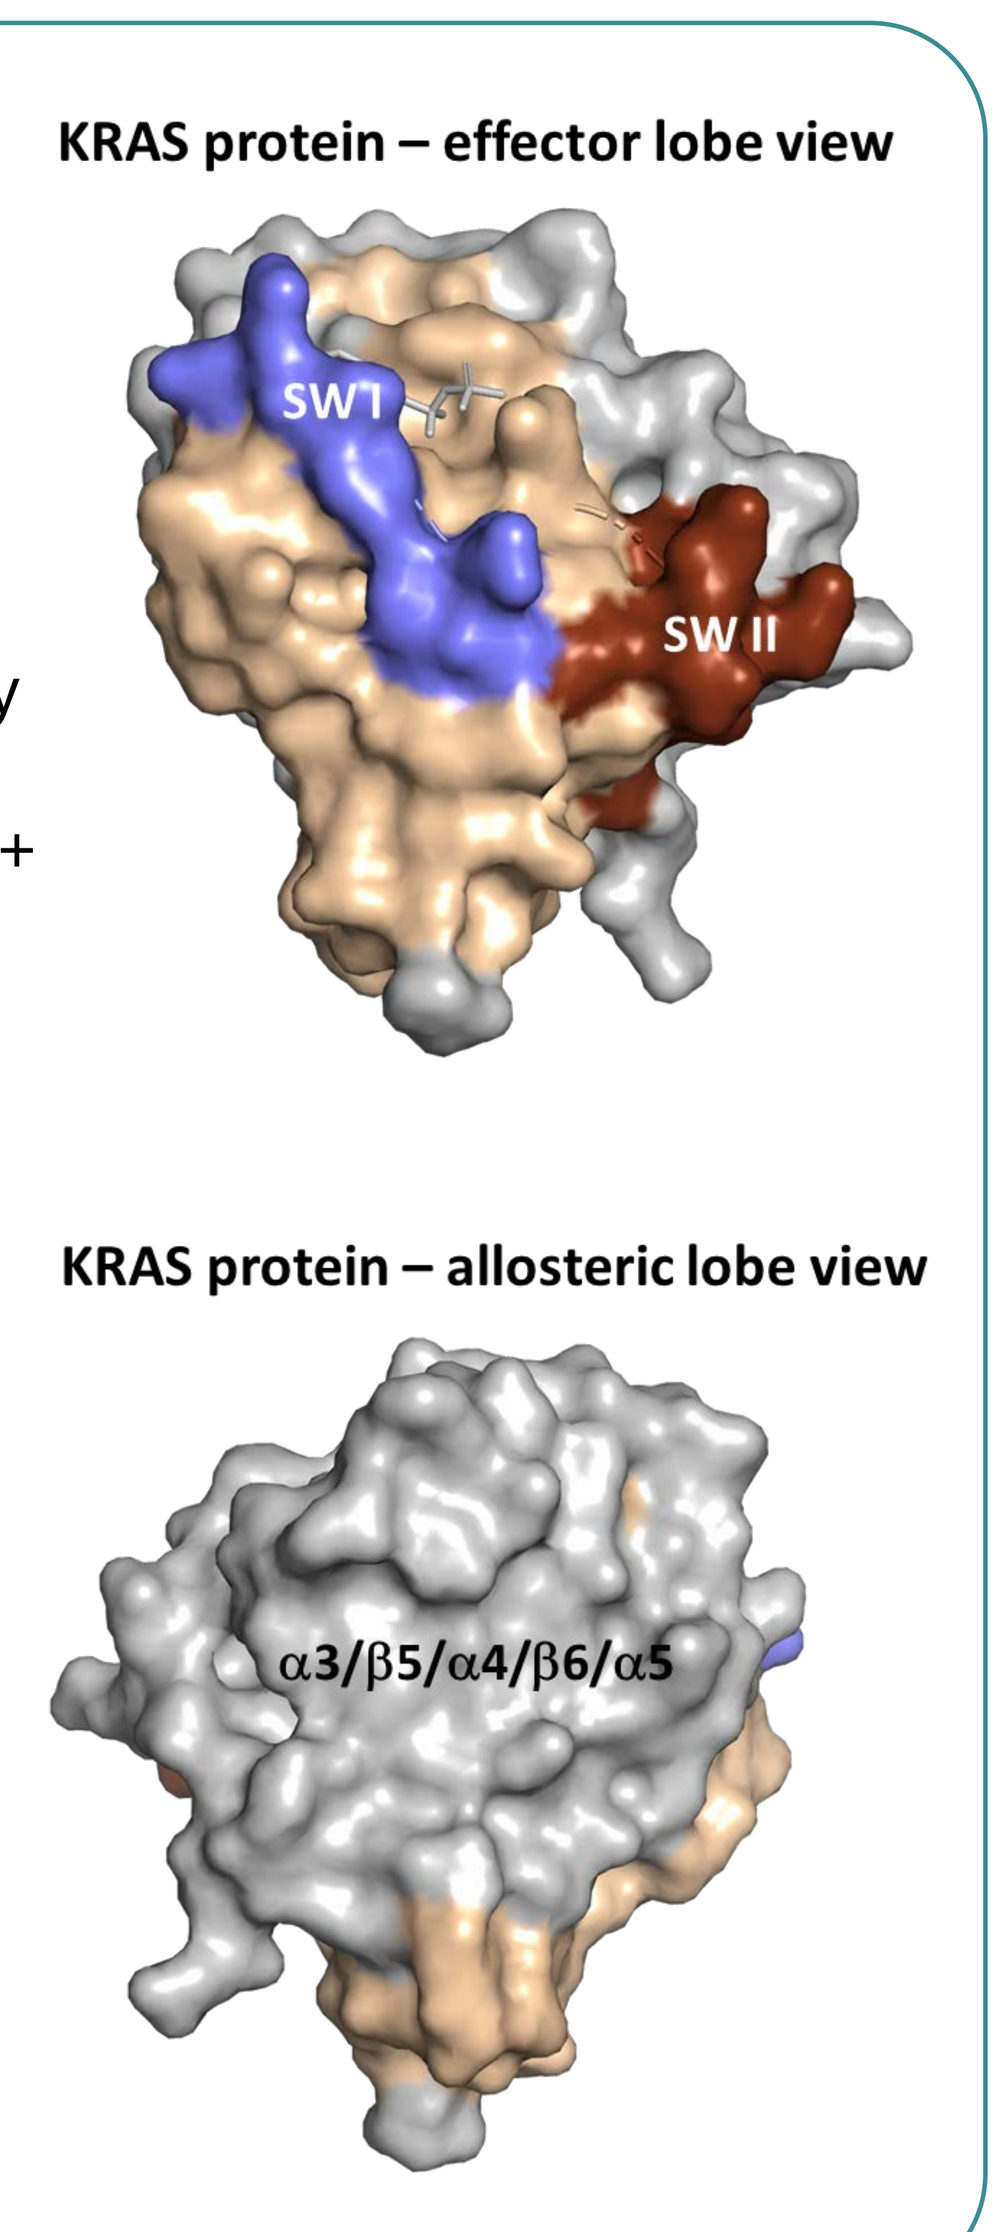

KRAS protein – effector lobe view
SW I
SW II
KRAS protein – allosteric lobe view
α3/β5/α4/β6/α5

1) K19 binds KRAS allosteric lobe

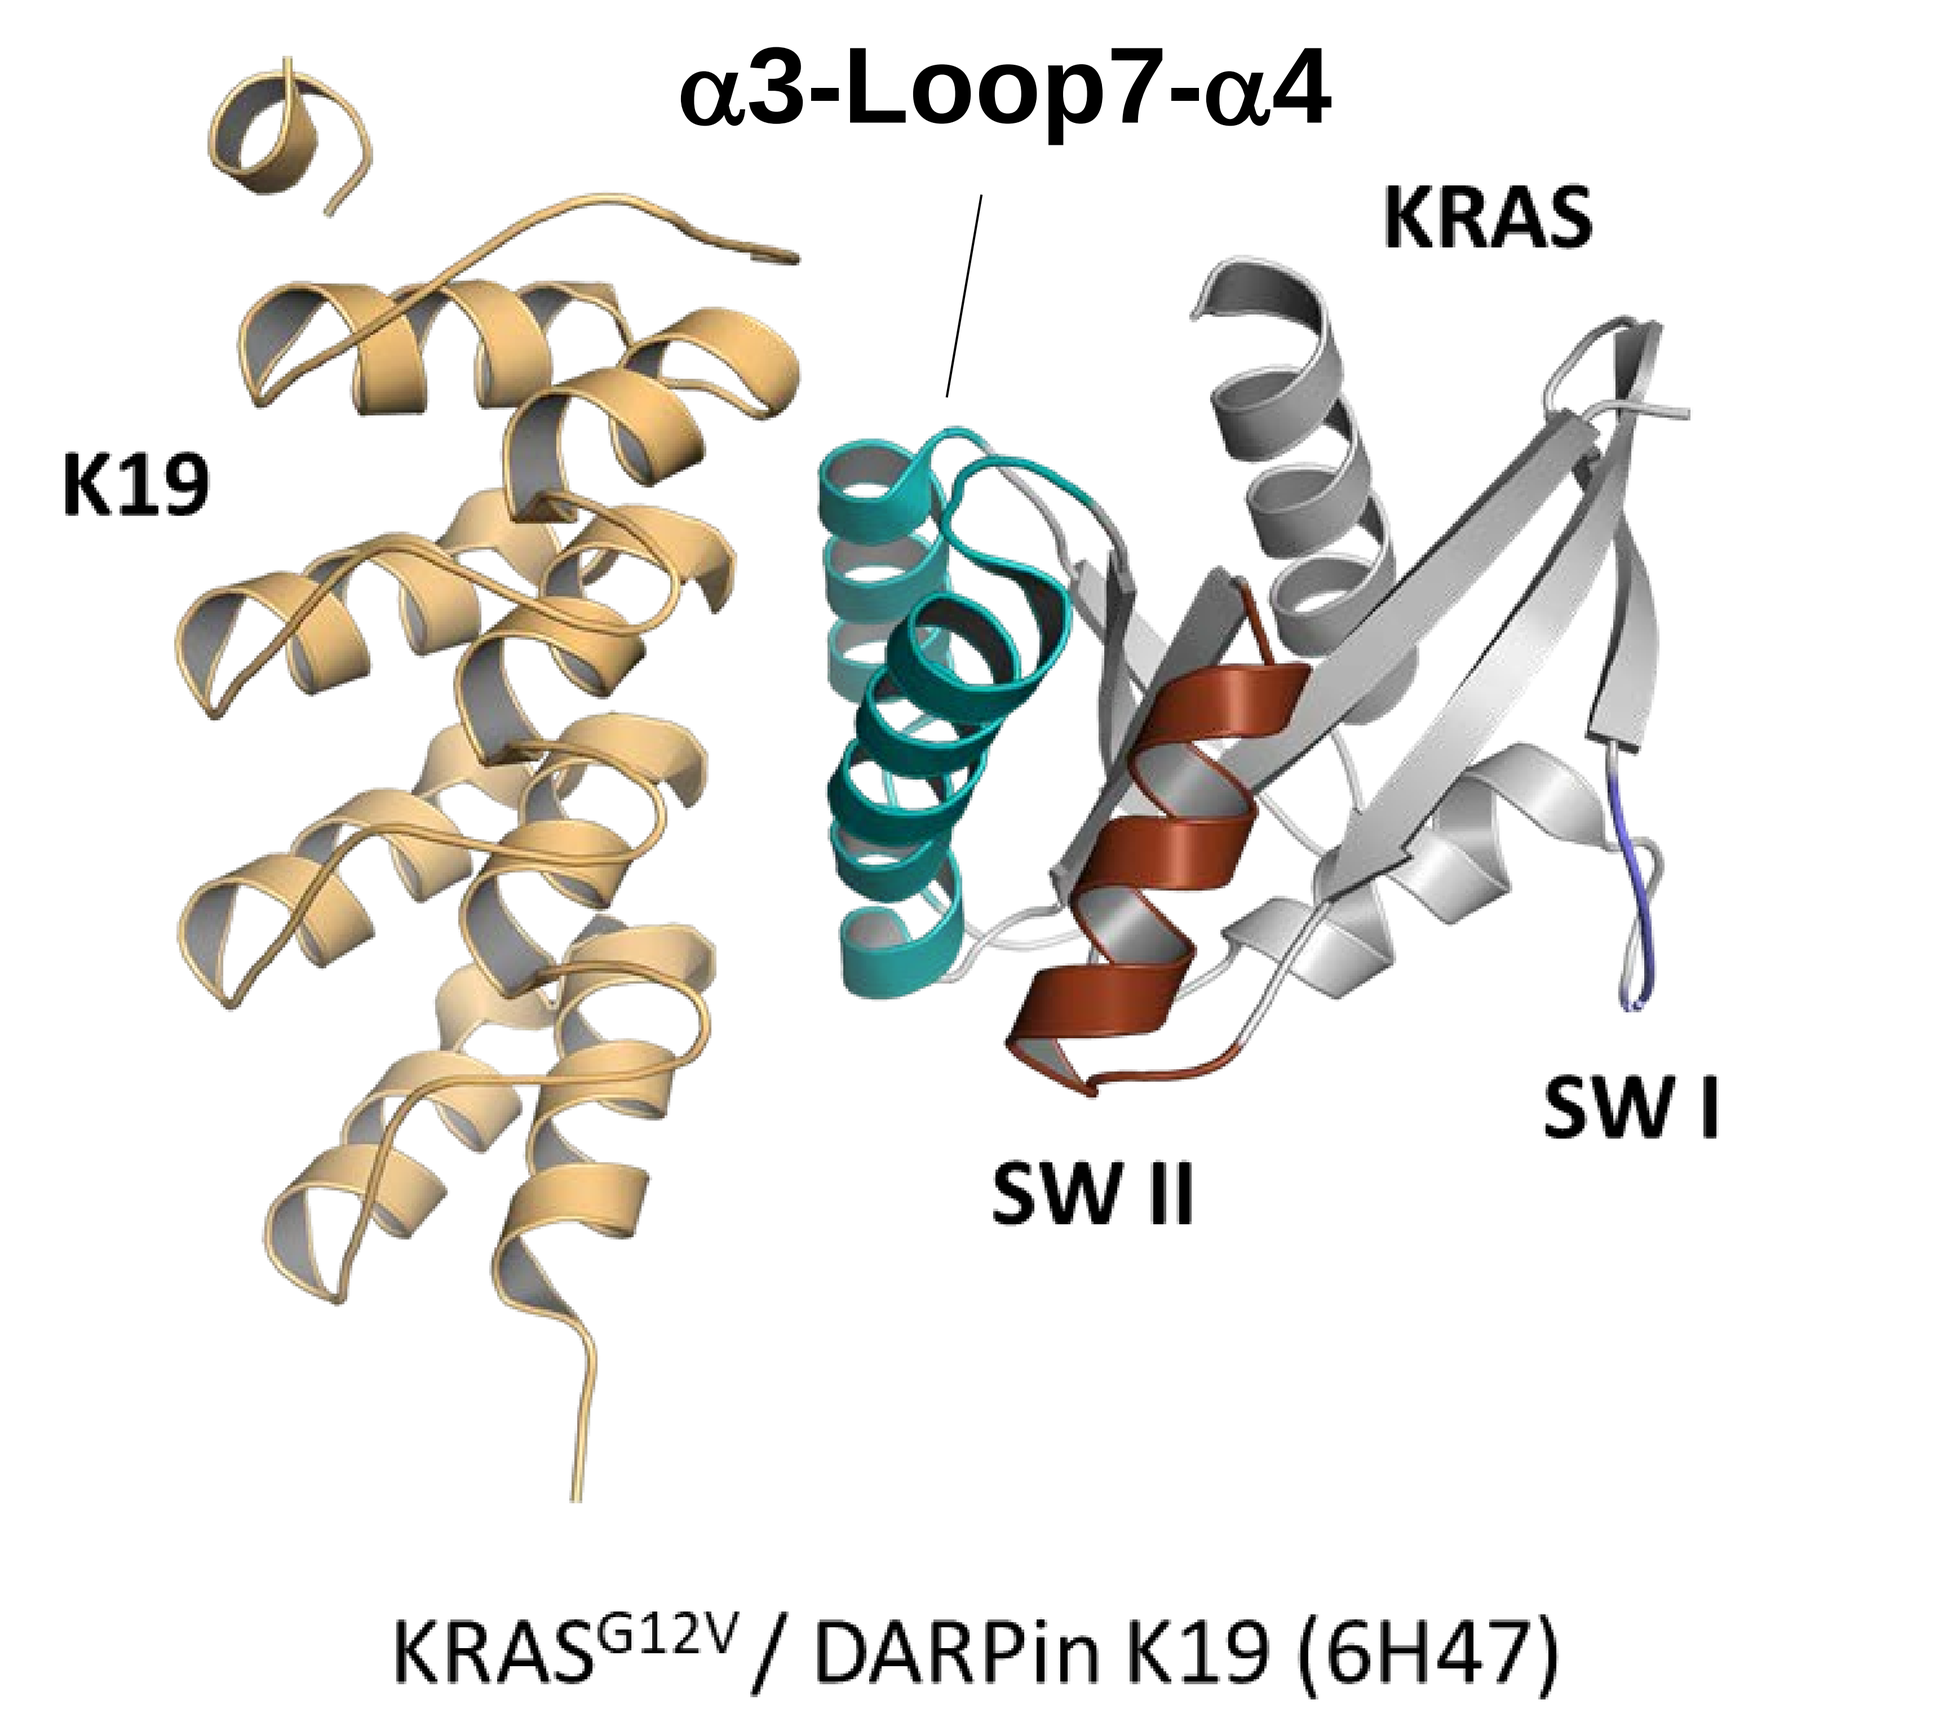

α3-Loop7-α4
KRAS
K19
SW II
SW I

$KRAS^{G12V}$ / DARPin K19 (6H47)

K_d ≈ 10nM for KRAS (-GDP and -GTP bound)

2) K19 binds specifically KRAS through its interaction with H95

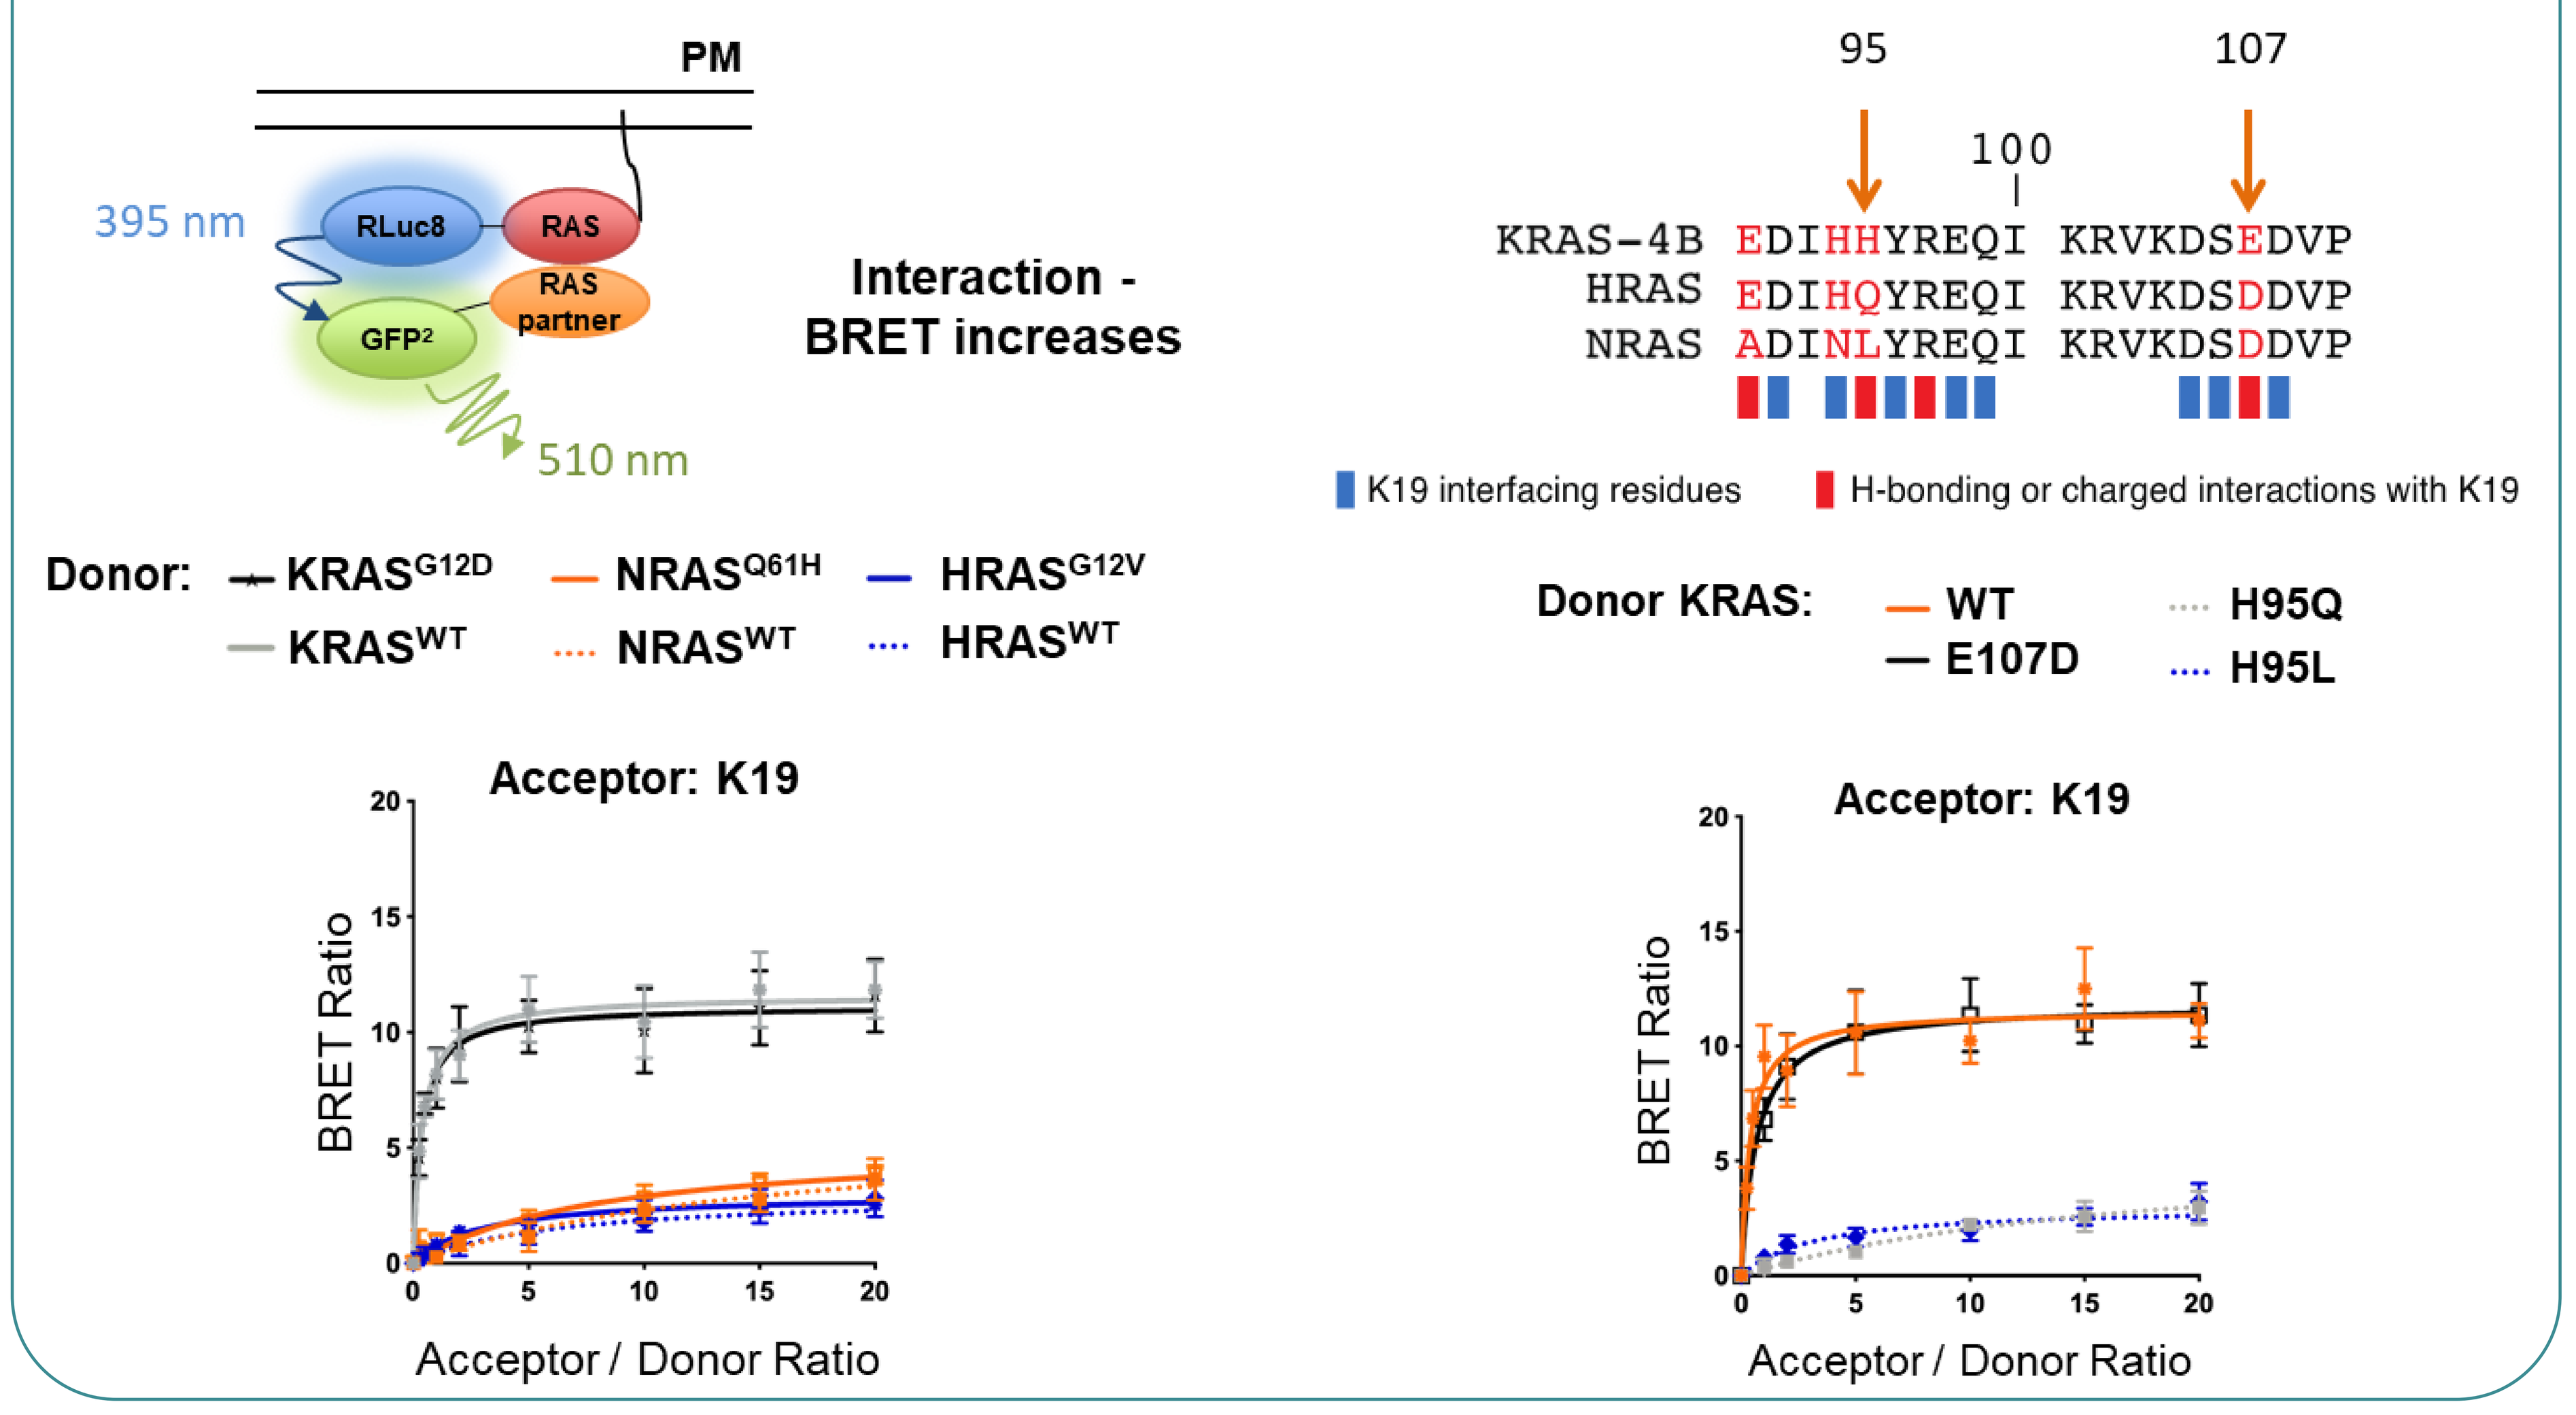

PM
395 nm
RLuc8
RAS
RAS partner
GFP2
510 nm
Interaction - BRET increases
Donor: KRASG12D NRASQ61H HRASG12V KRASWT NRASWT HRASWT
Acceptor: K19
BRET Ratio
Acceptor / Donor Ratio
95 100 107
KRAS-4B EDIHHYREQI KRVKDSEDVP
HRAS EDIHQYREQI KRVKDSDDVP
NRAS ADINLYREQI KRVKDSDDVP
K19 interfacing residues
H-bonding or charged interactions with K19
Donor KRAS: WT E107D H95Q H95L
Acceptor: K19
BRET Ratio
Acceptor / Donor Ratio

3) Development and characterisation of pan-RAS and KRAS-specific macromolecule degraders

a) Single domain-based targeted-protein degradation

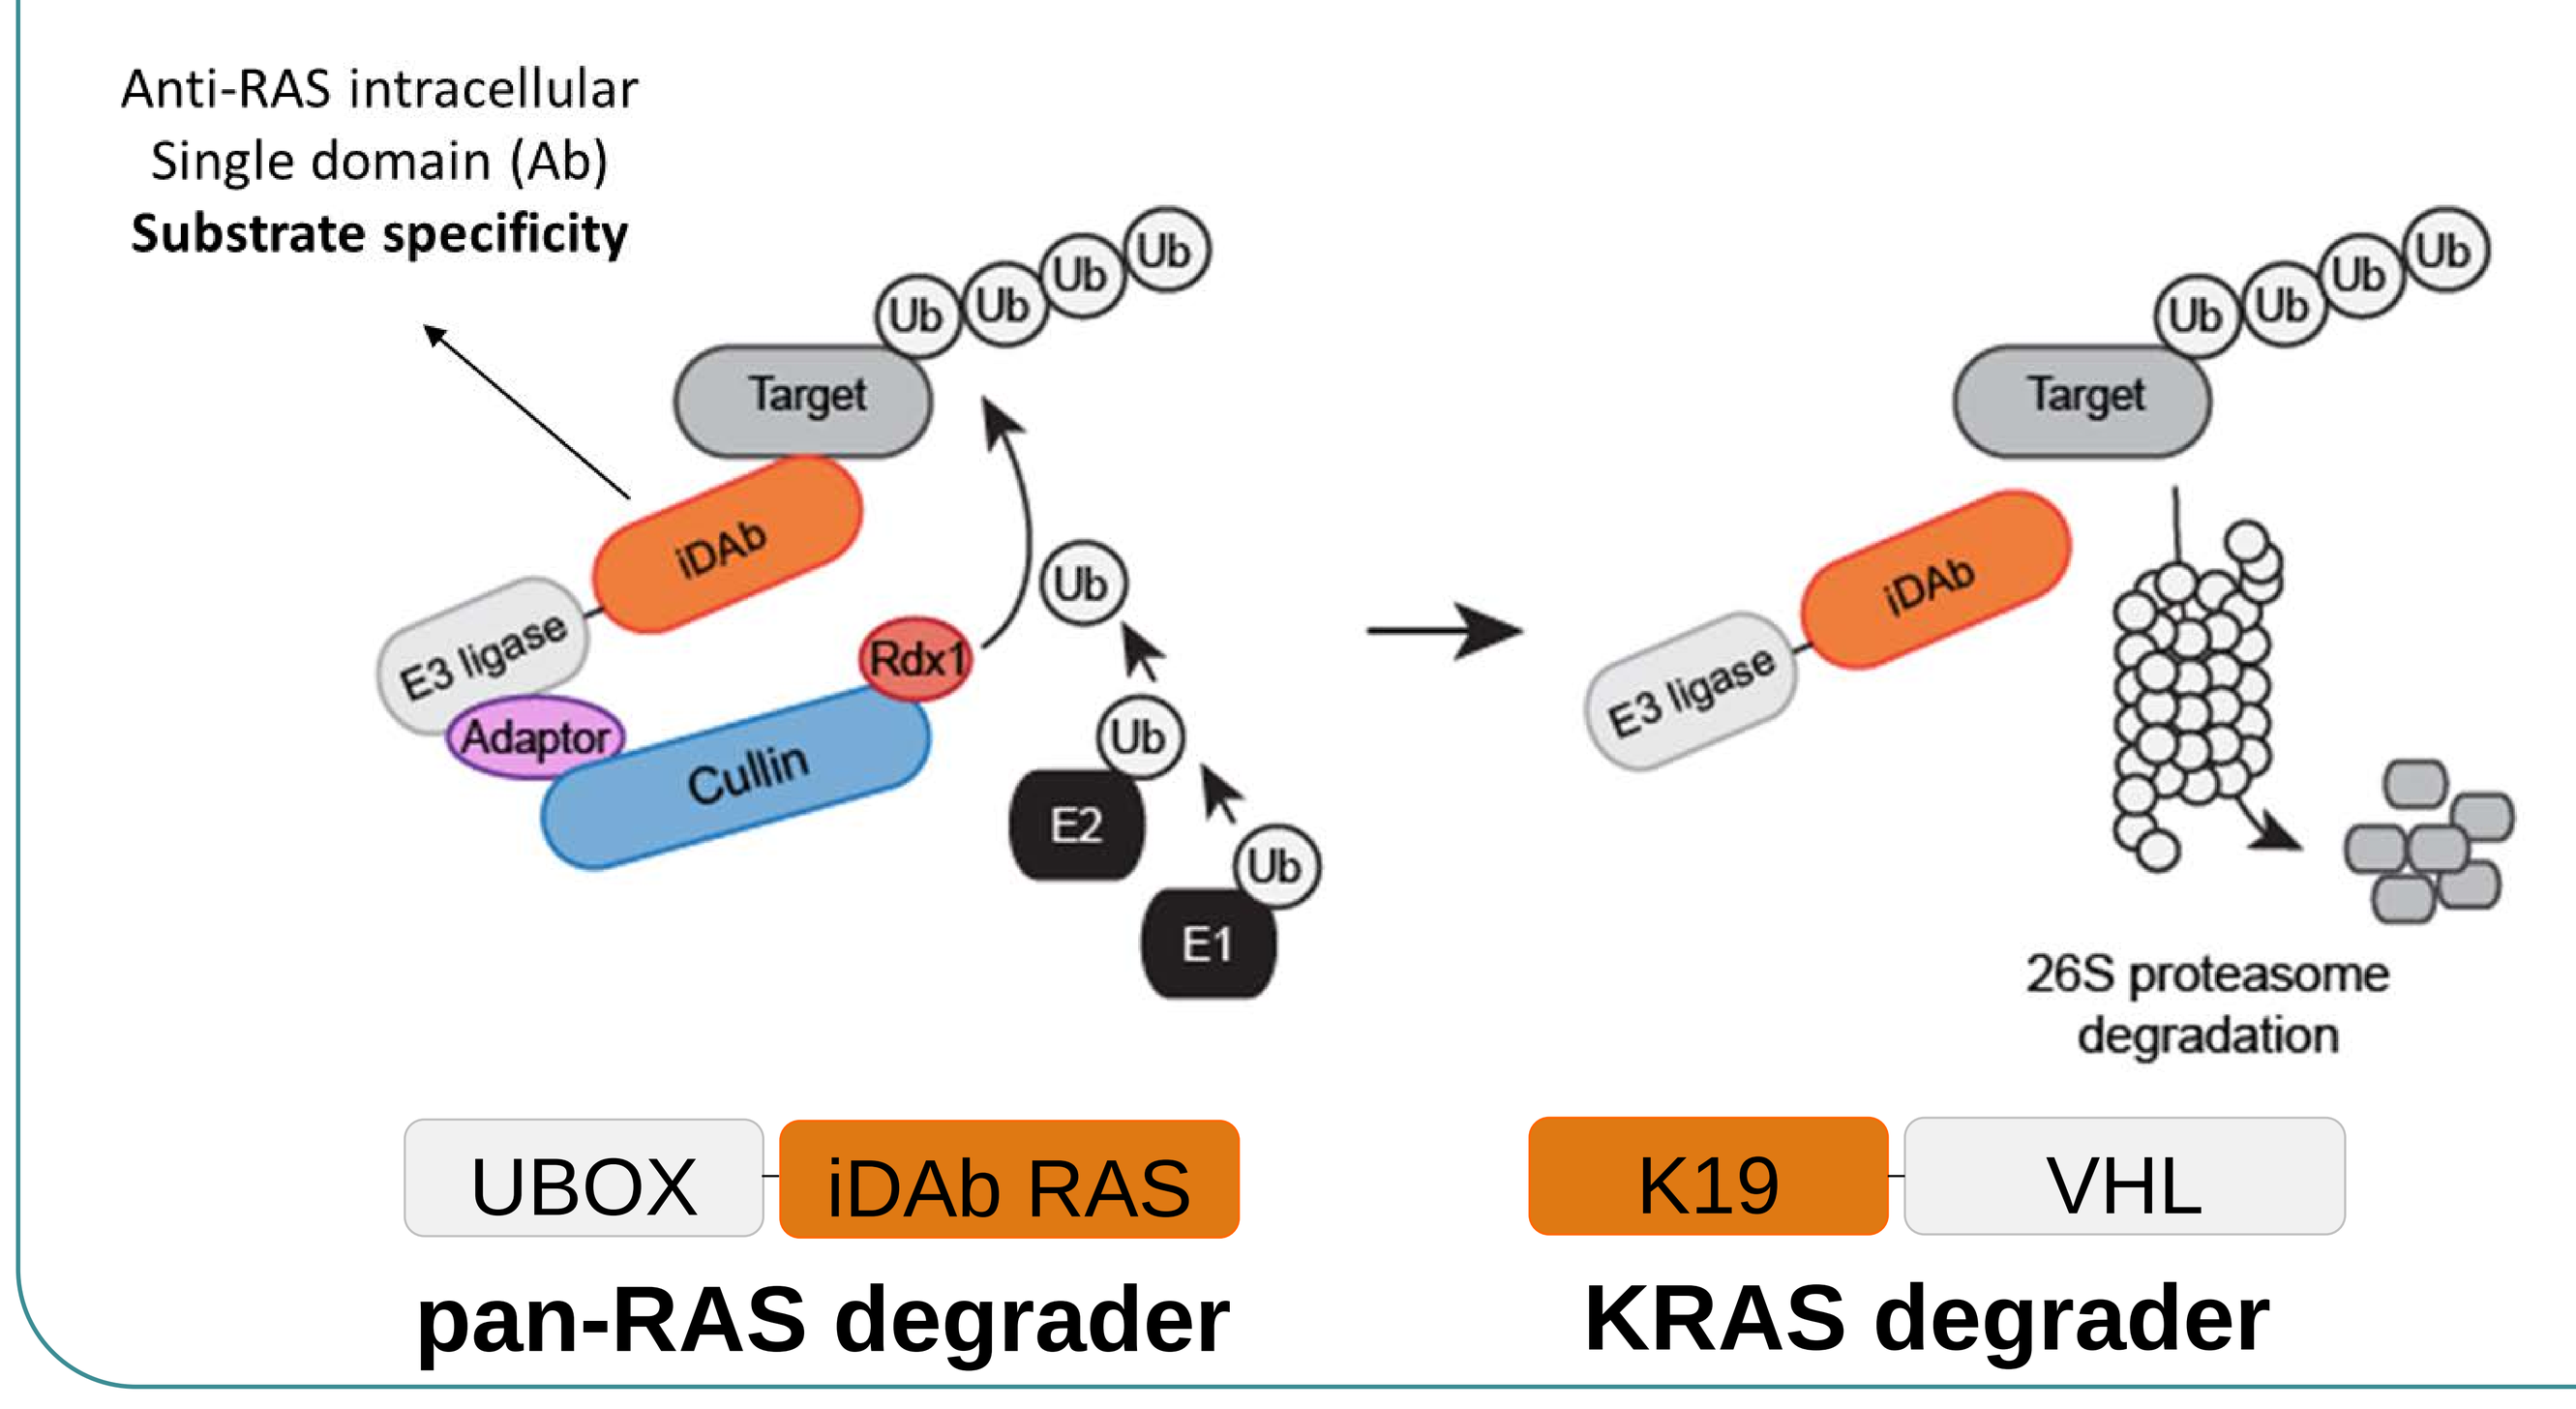

Anti-RAS intracellular
Single domain (Ab)
Substrate specificity
Target
Ub
iDAb
E3 ligase
Adaptor
Cullin
Rdx1
E2
E1
26S proteasome degradation
UBOX
iDAb RAS
K19
VHL
pan-RAS degrader
KRAS degrader

b) Efficient and specific depletion of endogenous RAS in cells

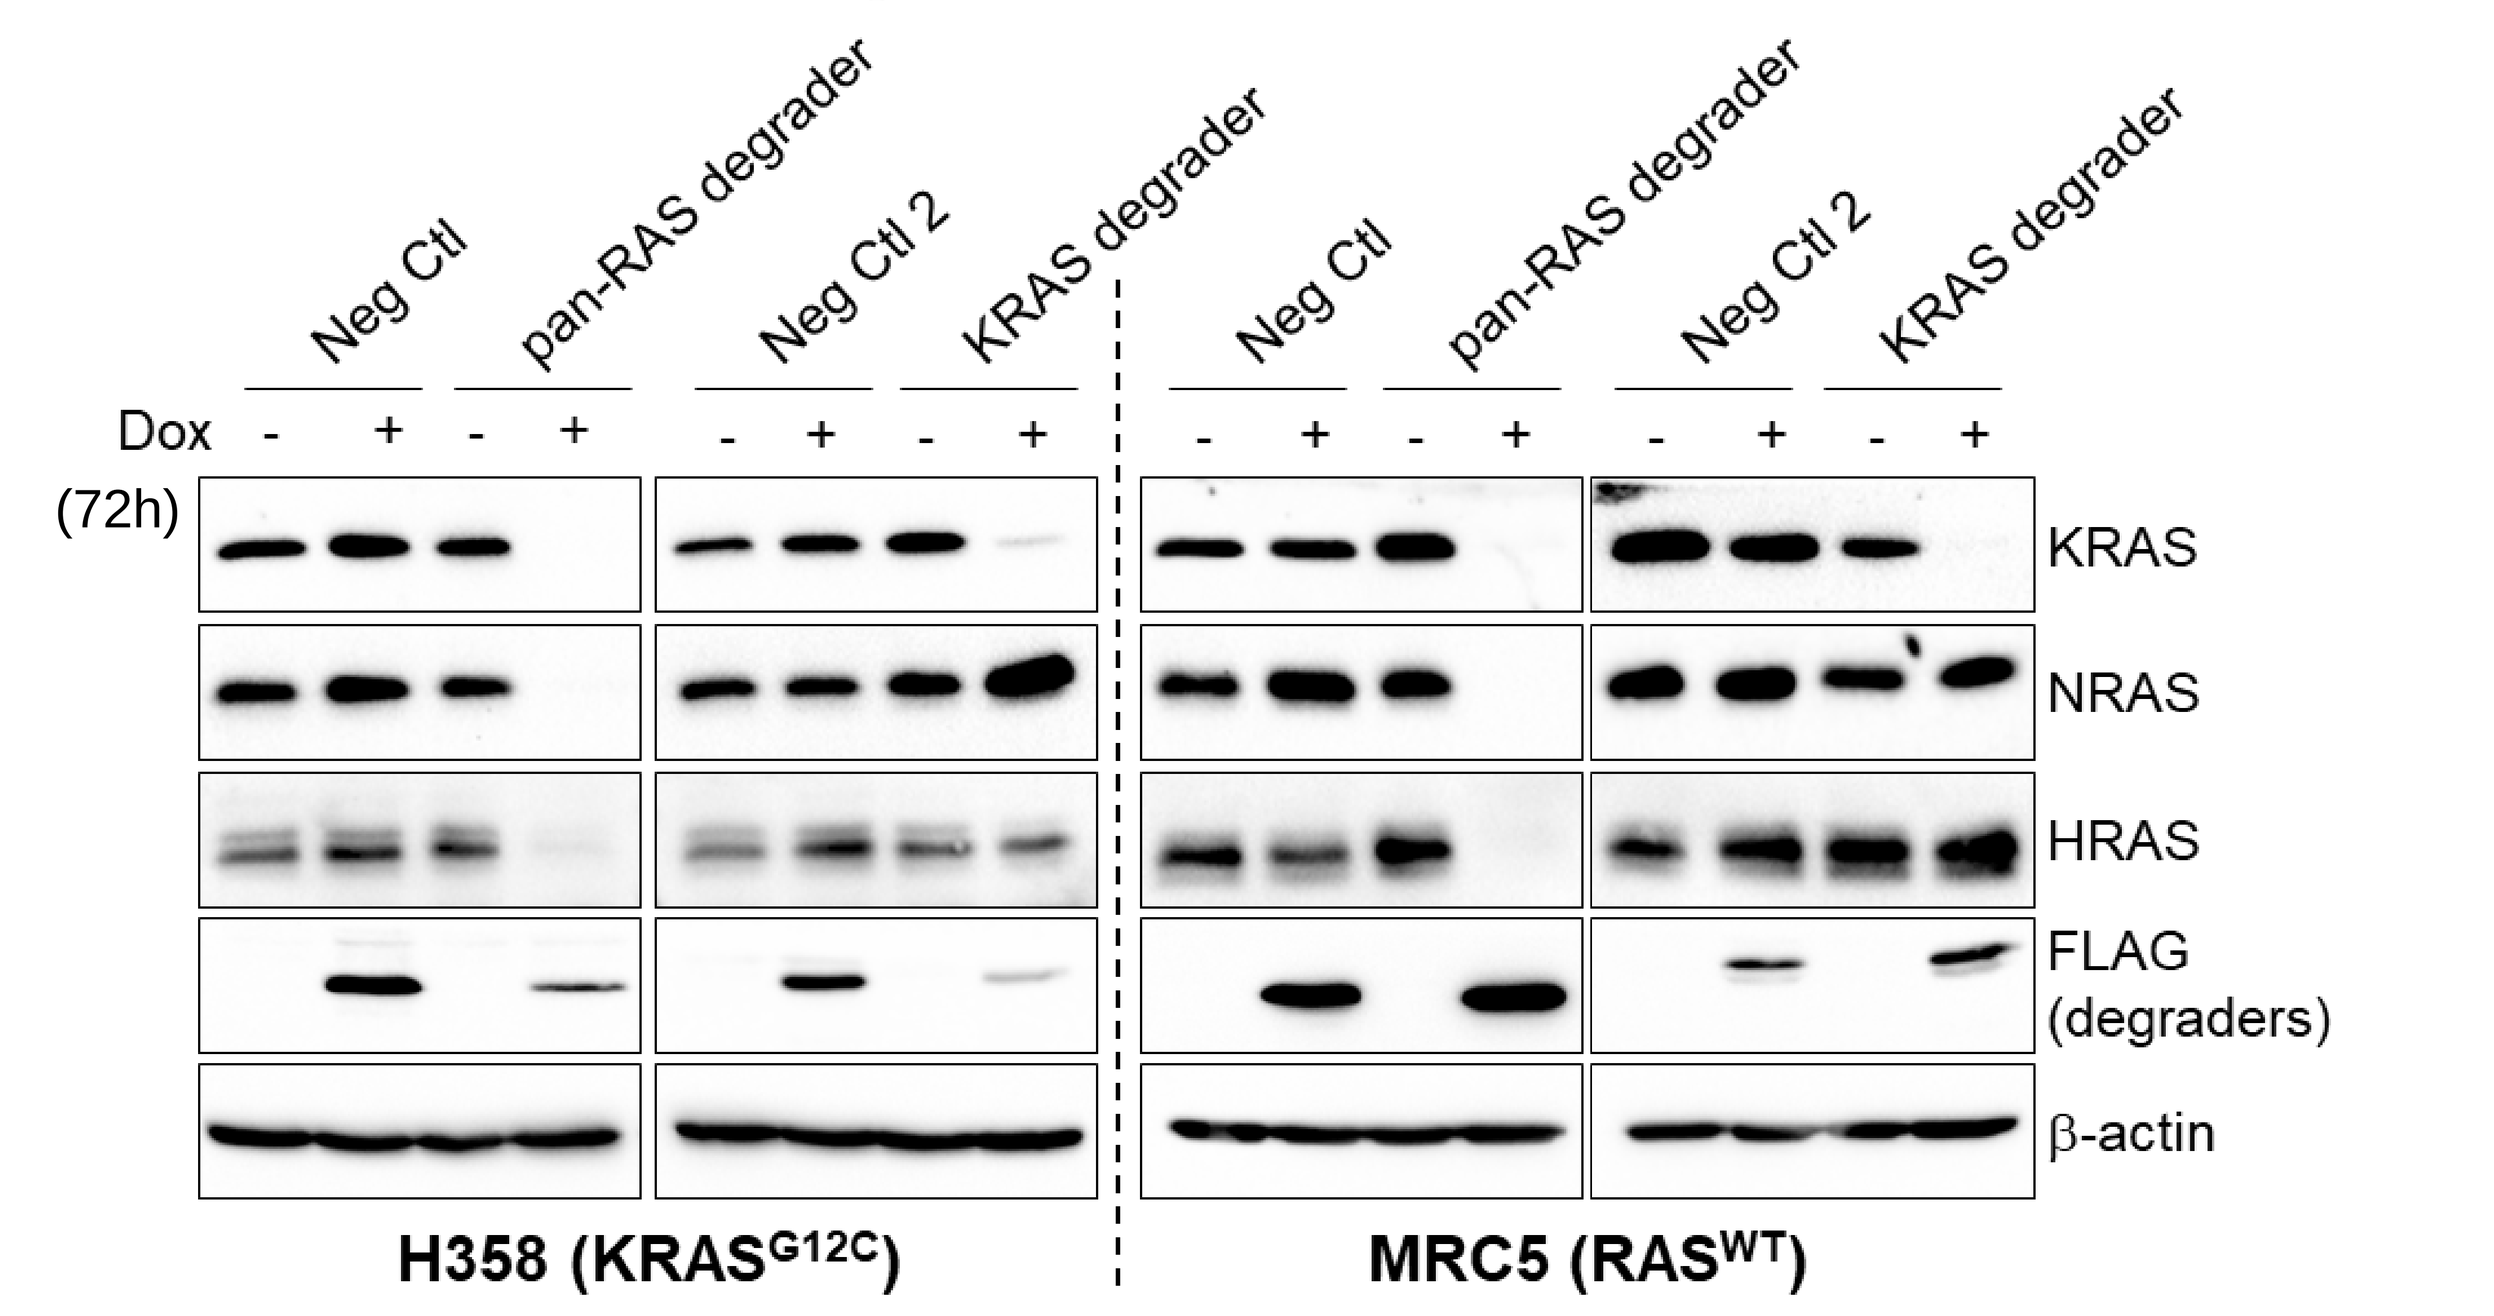

Neg Ctl
pan-RAS degrader
Neg Ctl 2
KRAS degrader
Dox
(72h)
KRAS
NRAS
HRAS
FLAG (degraders)
β-actin
H358 (KRASG12C)
MRC5 (RASWT)

c) Fast, sustained and specific degradation

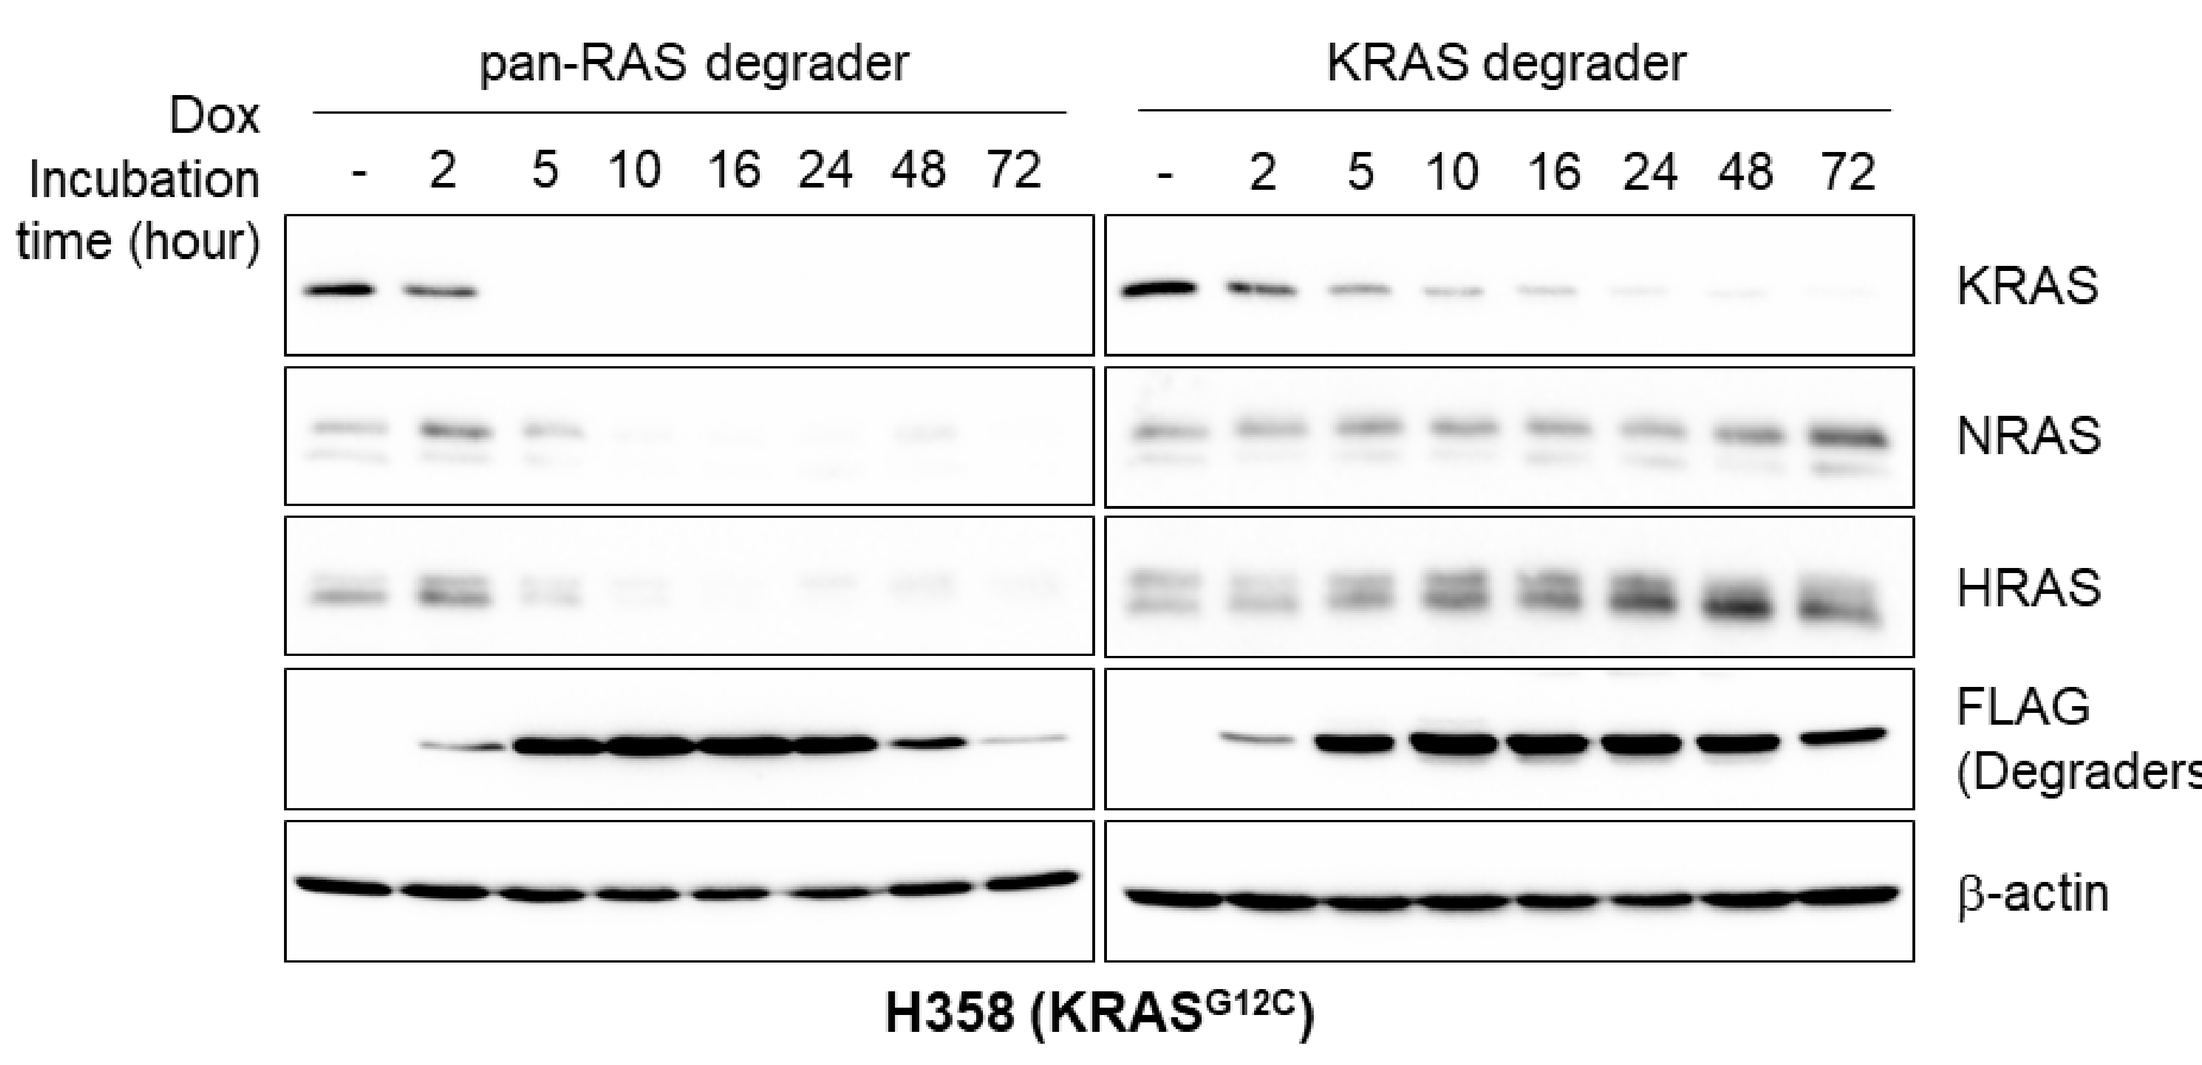

pan-RAS degrader
KRAS degrader
Dox Incubation time (hour)
- 2 5 10 16 24 48 72
KRAS
NRAS
HRAS
FLAG (Degraders)
β-actin
H358 (KRASG12C)

4) Specific inhibition of mutant KRAS cancer cells and tumours with KRAS degrader

a) KRAS degrader impairs RAS signalling specifically in mutant KRAS cancer cells

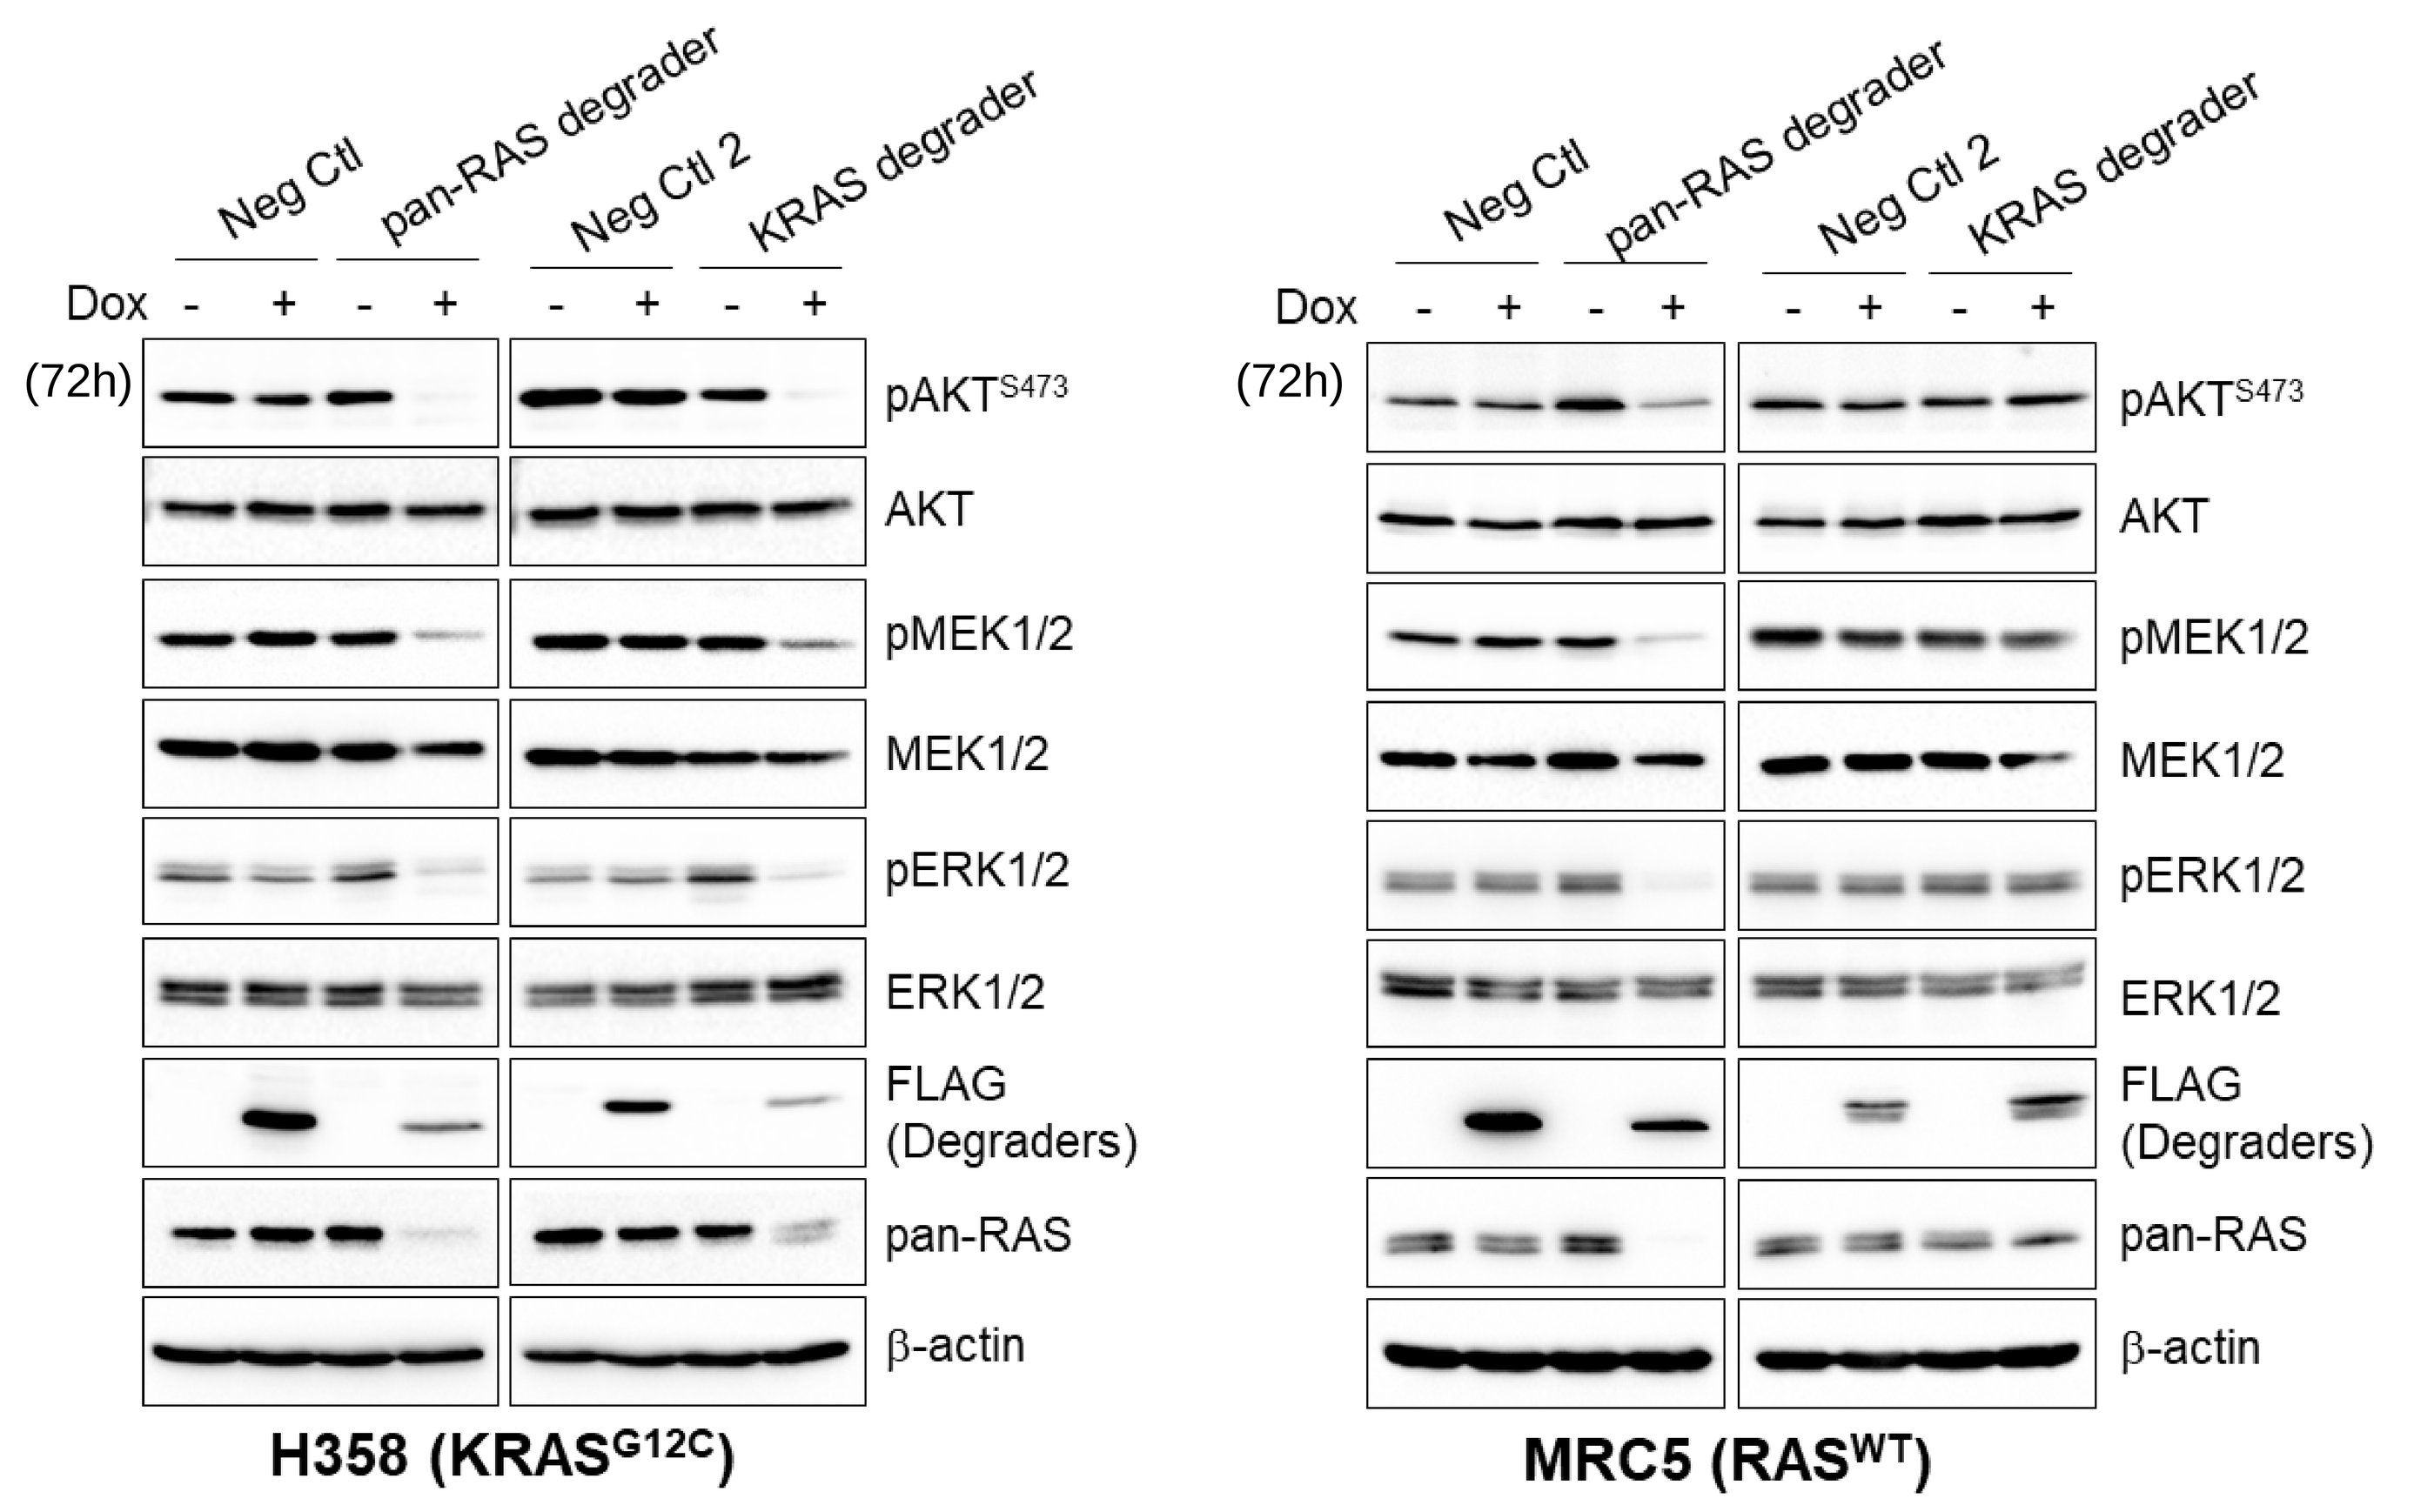

Neg Ctl
pan-RAS degrader
Neg Ctl 2
KRAS degrader
Dox
(72h)
pAKTS473
AKT
pMEK1/2
MEK1/2
pERK1/2
ERK1/2
FLAG (Degraders)
pan-RAS
β-actin
H358 (KRASG12C)
MRC5 (RASWT)

b) KRAS degrader inhibits specifically mutant KRAS cancer cells growth *in vitro*

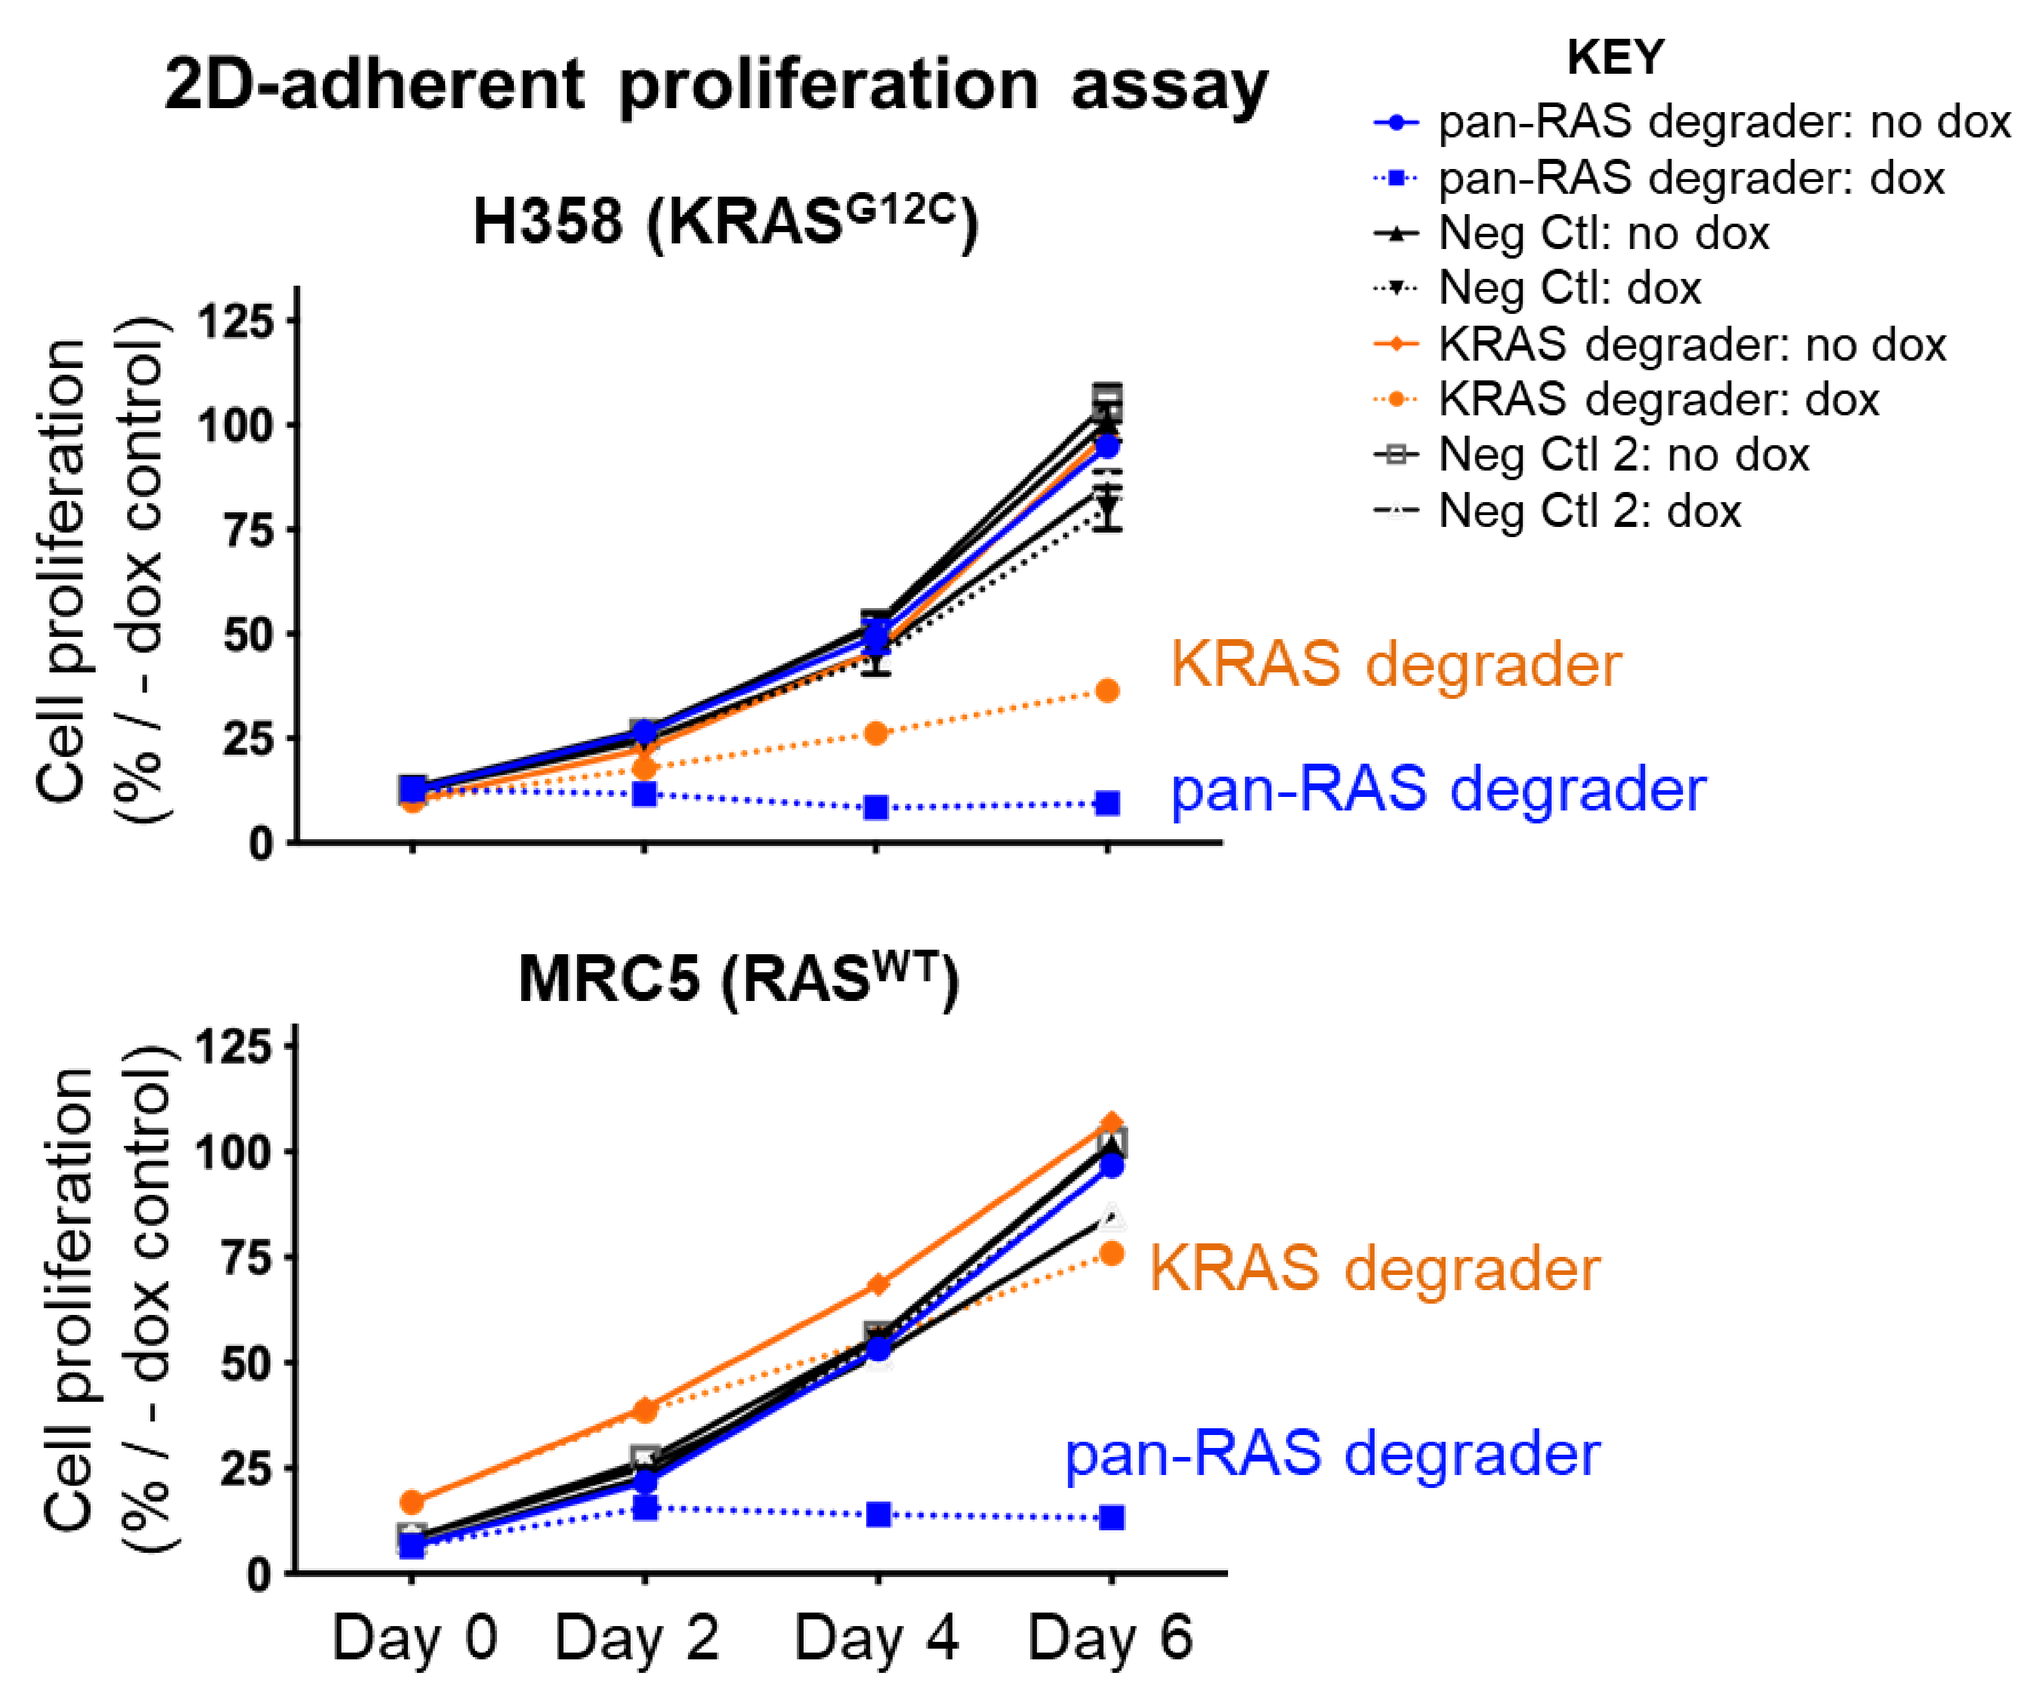

2D-adherent proliferation assay
H358 (KRASG12C)
KEY
pan-RAS degrader: no dox
pan-RAS degrader: dox
Neg Ctl: no dox
Neg Ctl: dox
KRAS degrader: no dox
KRAS degrader: dox
Neg Ctl 2: no dox
Neg Ctl 2: dox
Cell proliferation (% / - dox control)
KRAS degrader
pan-RAS degrader
MRC5 (RASWT)
Day 0 Day 2 Day 4 Day 6

c) KRAS degrader induces regression of mutant KRAS driven tumours *in vivo*

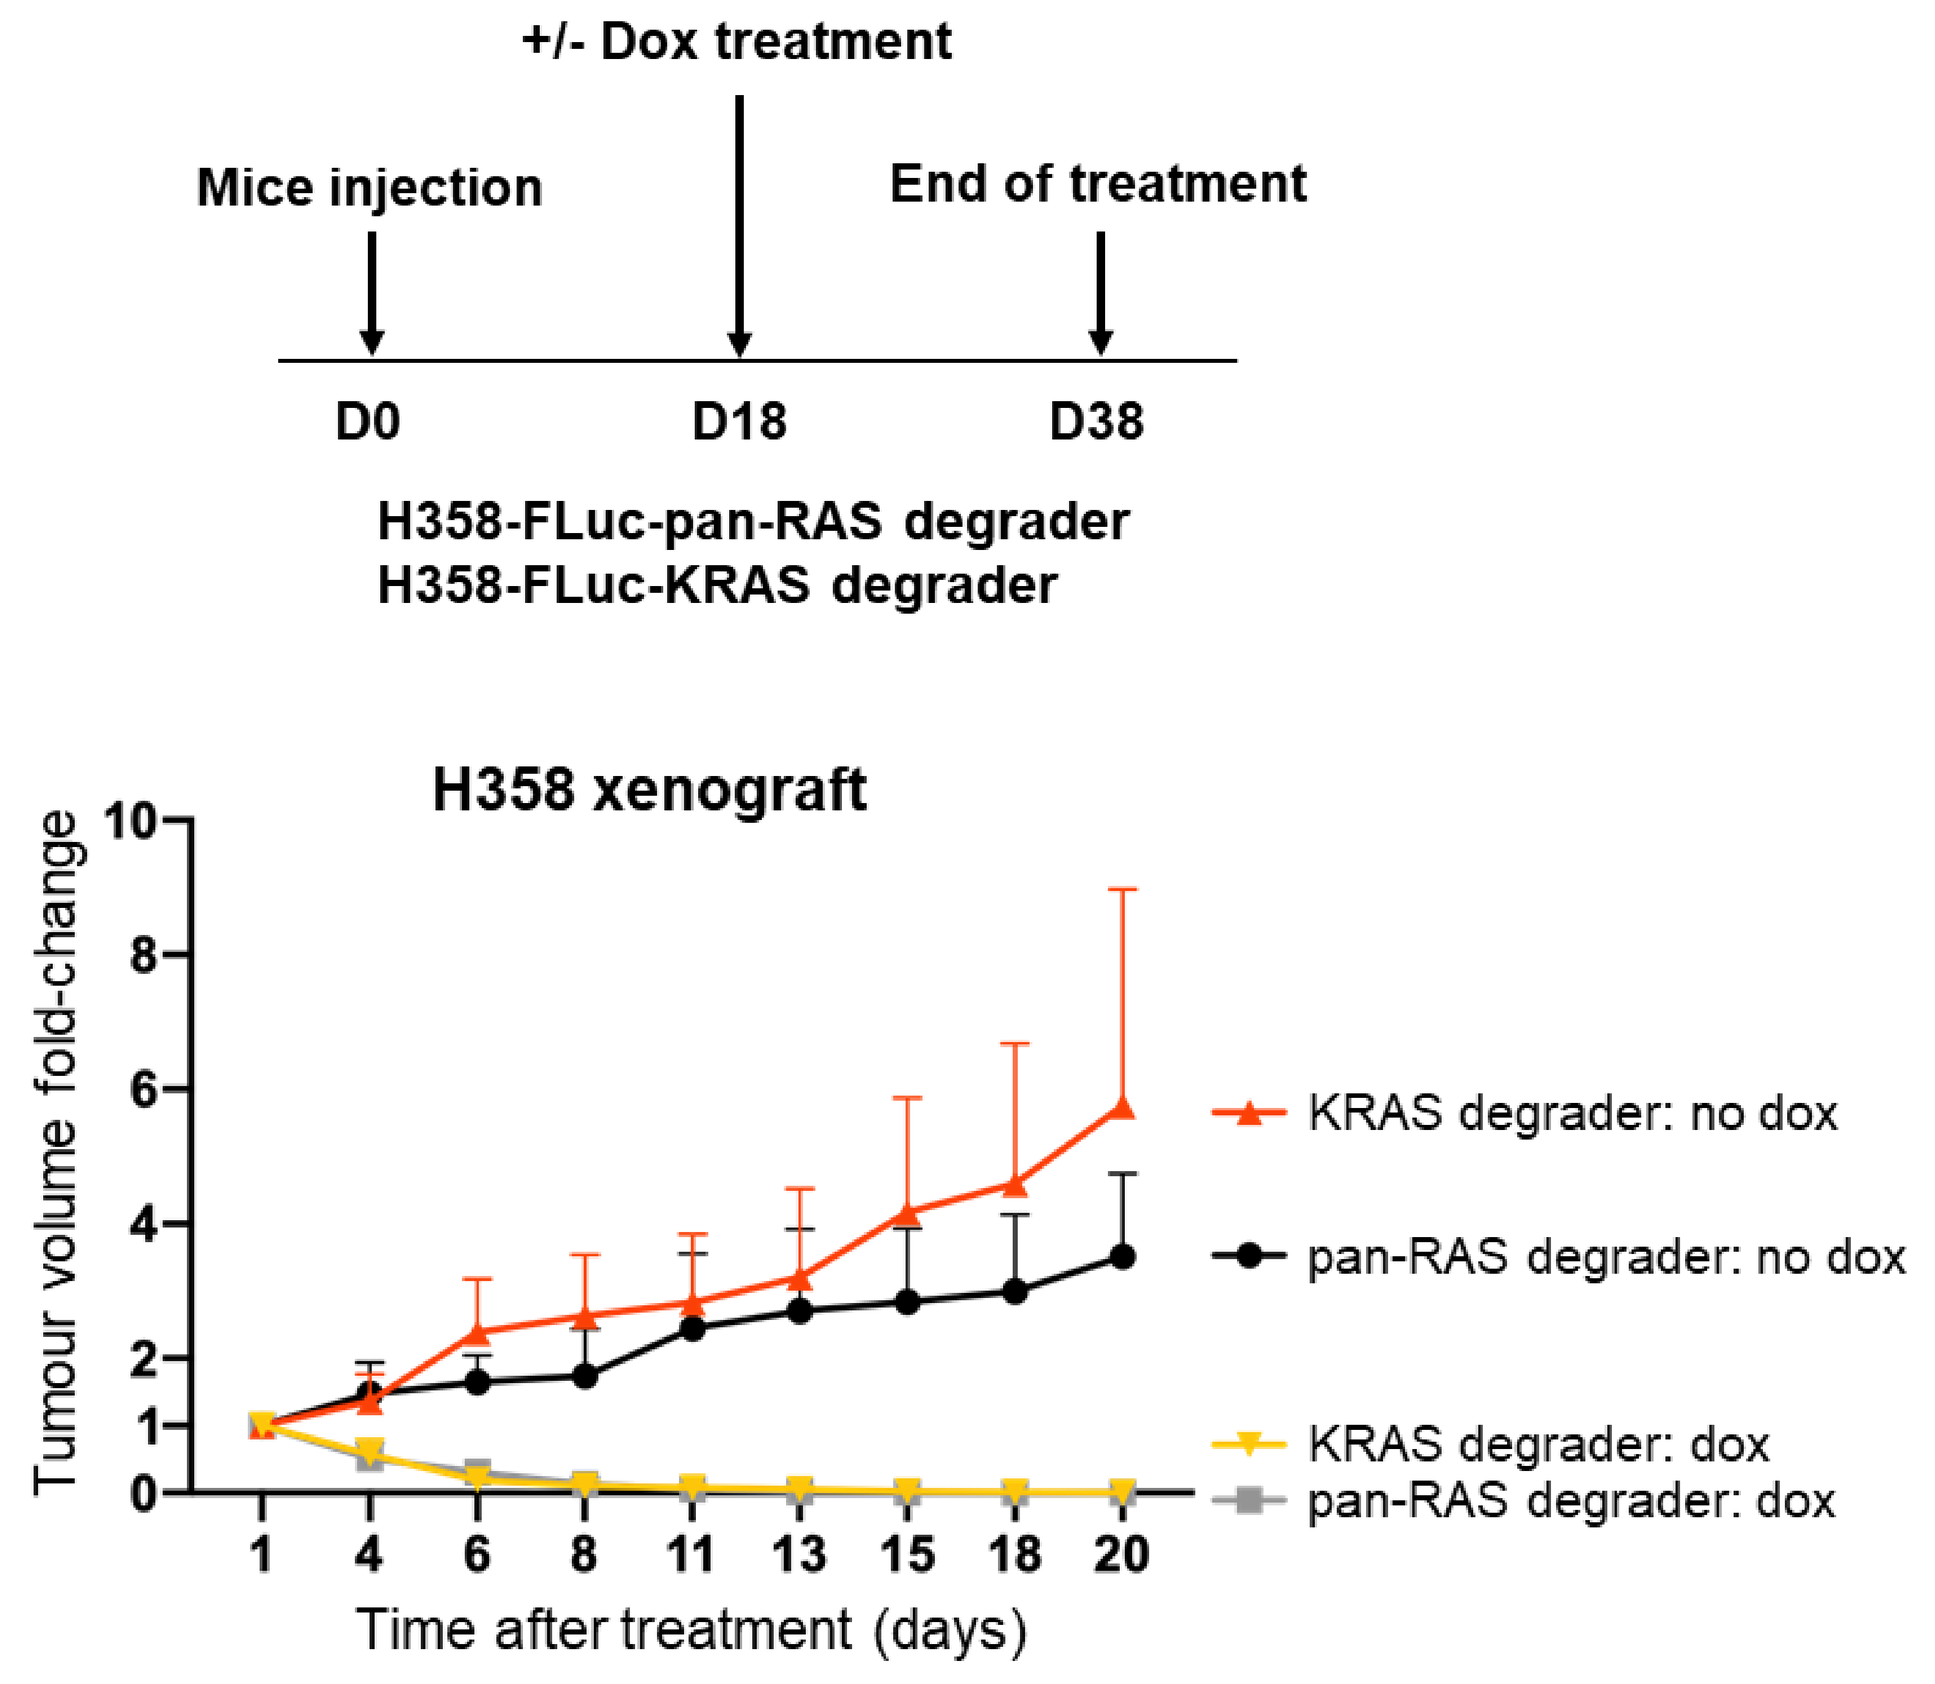

+/- Dox treatment
Mice injection
End of treatment
D0 D18 D38
H358-FLuc-pan-RAS degrader
H358-FLuc-KRAS degrader
H358 xenograft
Tumour volume fold-change
KRAS degrader: no dox
pan-RAS degrader: no dox
KRAS degrader: dox
pan-RAS degrader: dox
Time after treatment (days)

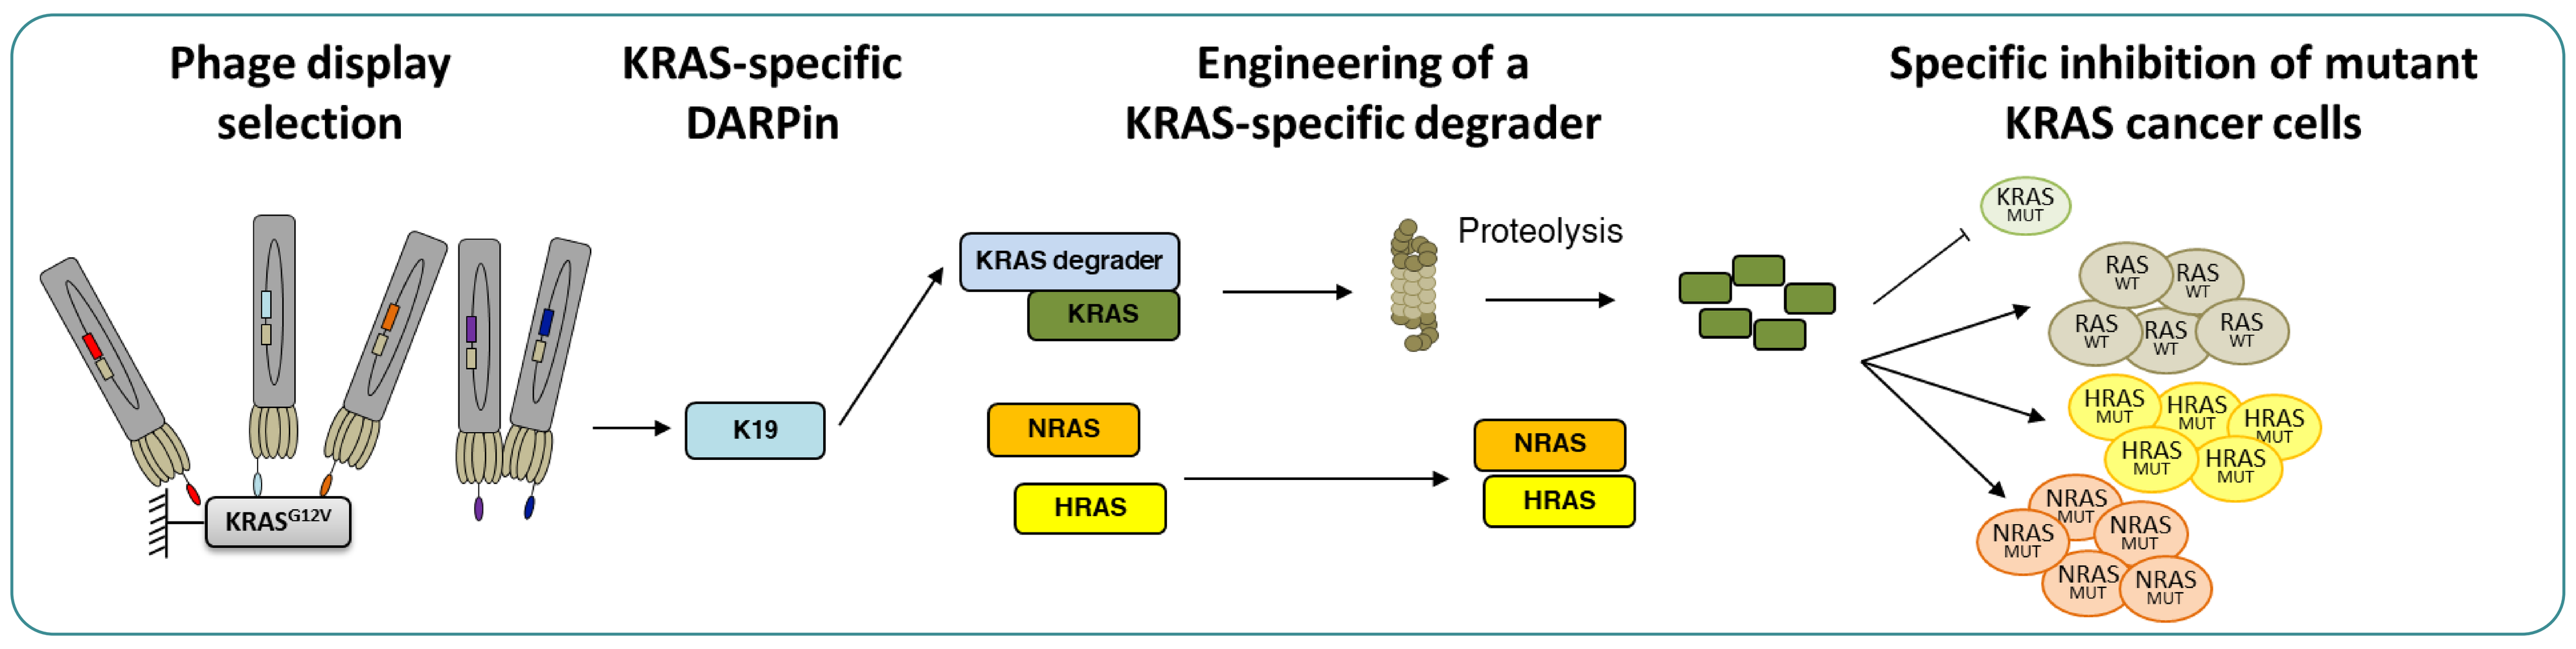

Phage display selection
KRAS-specific DARPin
Engineering of a KRAS-specific degrader
Specific inhibition of mutant KRAS cancer cells
KRASG12V
K19
KRAS degrader
KRAS
NRAS
HRAS
Proteolysis

This work was published in:
Bery N, et al. Nature Communications. 2019 (KRAS-specific DARPins)
Bery N, et al. Nature Communications. 2020 (macromolecule-based RAS/KRAS degraders)

The schematic diagram displayed in 3) was adapted from Bery N, et al. Cell Chem. Biol. 2019